Reaffirming Our Commitment to CPD: Advancing Interprofessional Continuing Education (IPCE) for All

BACKGROUND / INTRODUCTION

In **2022**, during preparation of a **Joint Accreditation self-study**, an organizational assessment was conducted to evaluate stakeholder understanding and valuing of i**nterprofessional collaborative practice (IPCP)** and **interprofessional continuing education (IPCE)**.

Interprofessional continuing education has been shown to improve collaboration and patient outcomes; however, fewer studies examine whether **sustained implementation of IPCE** contributes to **organizational culture change** within accredited continuing professional development (CPD) systems.

This study evaluates l**ongitudinal changes in stakeholder perceptions** following implementation of IPCE-aligned educational practices.

RESEARCH QUESTION / PURPOSE

This study examined whether **sustained implementation of interprofessional continuing education (IPCE)** influenced stakeholder attitudes and knowledge over time and whether these concepts extended beyond formal continuing education stakeholders.

Specifically, the study asked:

- **Attitudes and Knowledge:**
 To what extent does sustained implementation of interprofessional continuing education (IPCE) influence stakeholder* attitudes and knowledge from baseline (T1: 2022 assessment) to follow-up (T2: 2025 assessment)?
- **Broader Professional Environment:**
 To what extent are interprofessional collaborative practice concepts evident among professionals beyond formal continuing education stakeholders at T2?

*Stakeholders include course directors, planning committee members, presenters, and learners participating in accredited continuing education activities.

METHODS

A longitudinal cohort comparison was conducted using a **2022 baseline organizational assessment** and a **2025 follow-up survey** distributed to identified continuing education stakeholders, including course directors, planning committee members, presenters, and learners.

The survey instrument was aligned across cohorts to allow **direct comparison over time**. **Descriptive statistics** were used to evaluate trends in stakeholder responses.

A **secondary anonymous survey** using the same instrument was administered during educational and networking events to explore broader professional exposure to interprofessional continuing education concepts beyond formal program planning roles.

The study was informed by the **Modified Kirkpatrick Model for Outcomes Evaluation** and the **Interprofessional Learning Continuum model**.

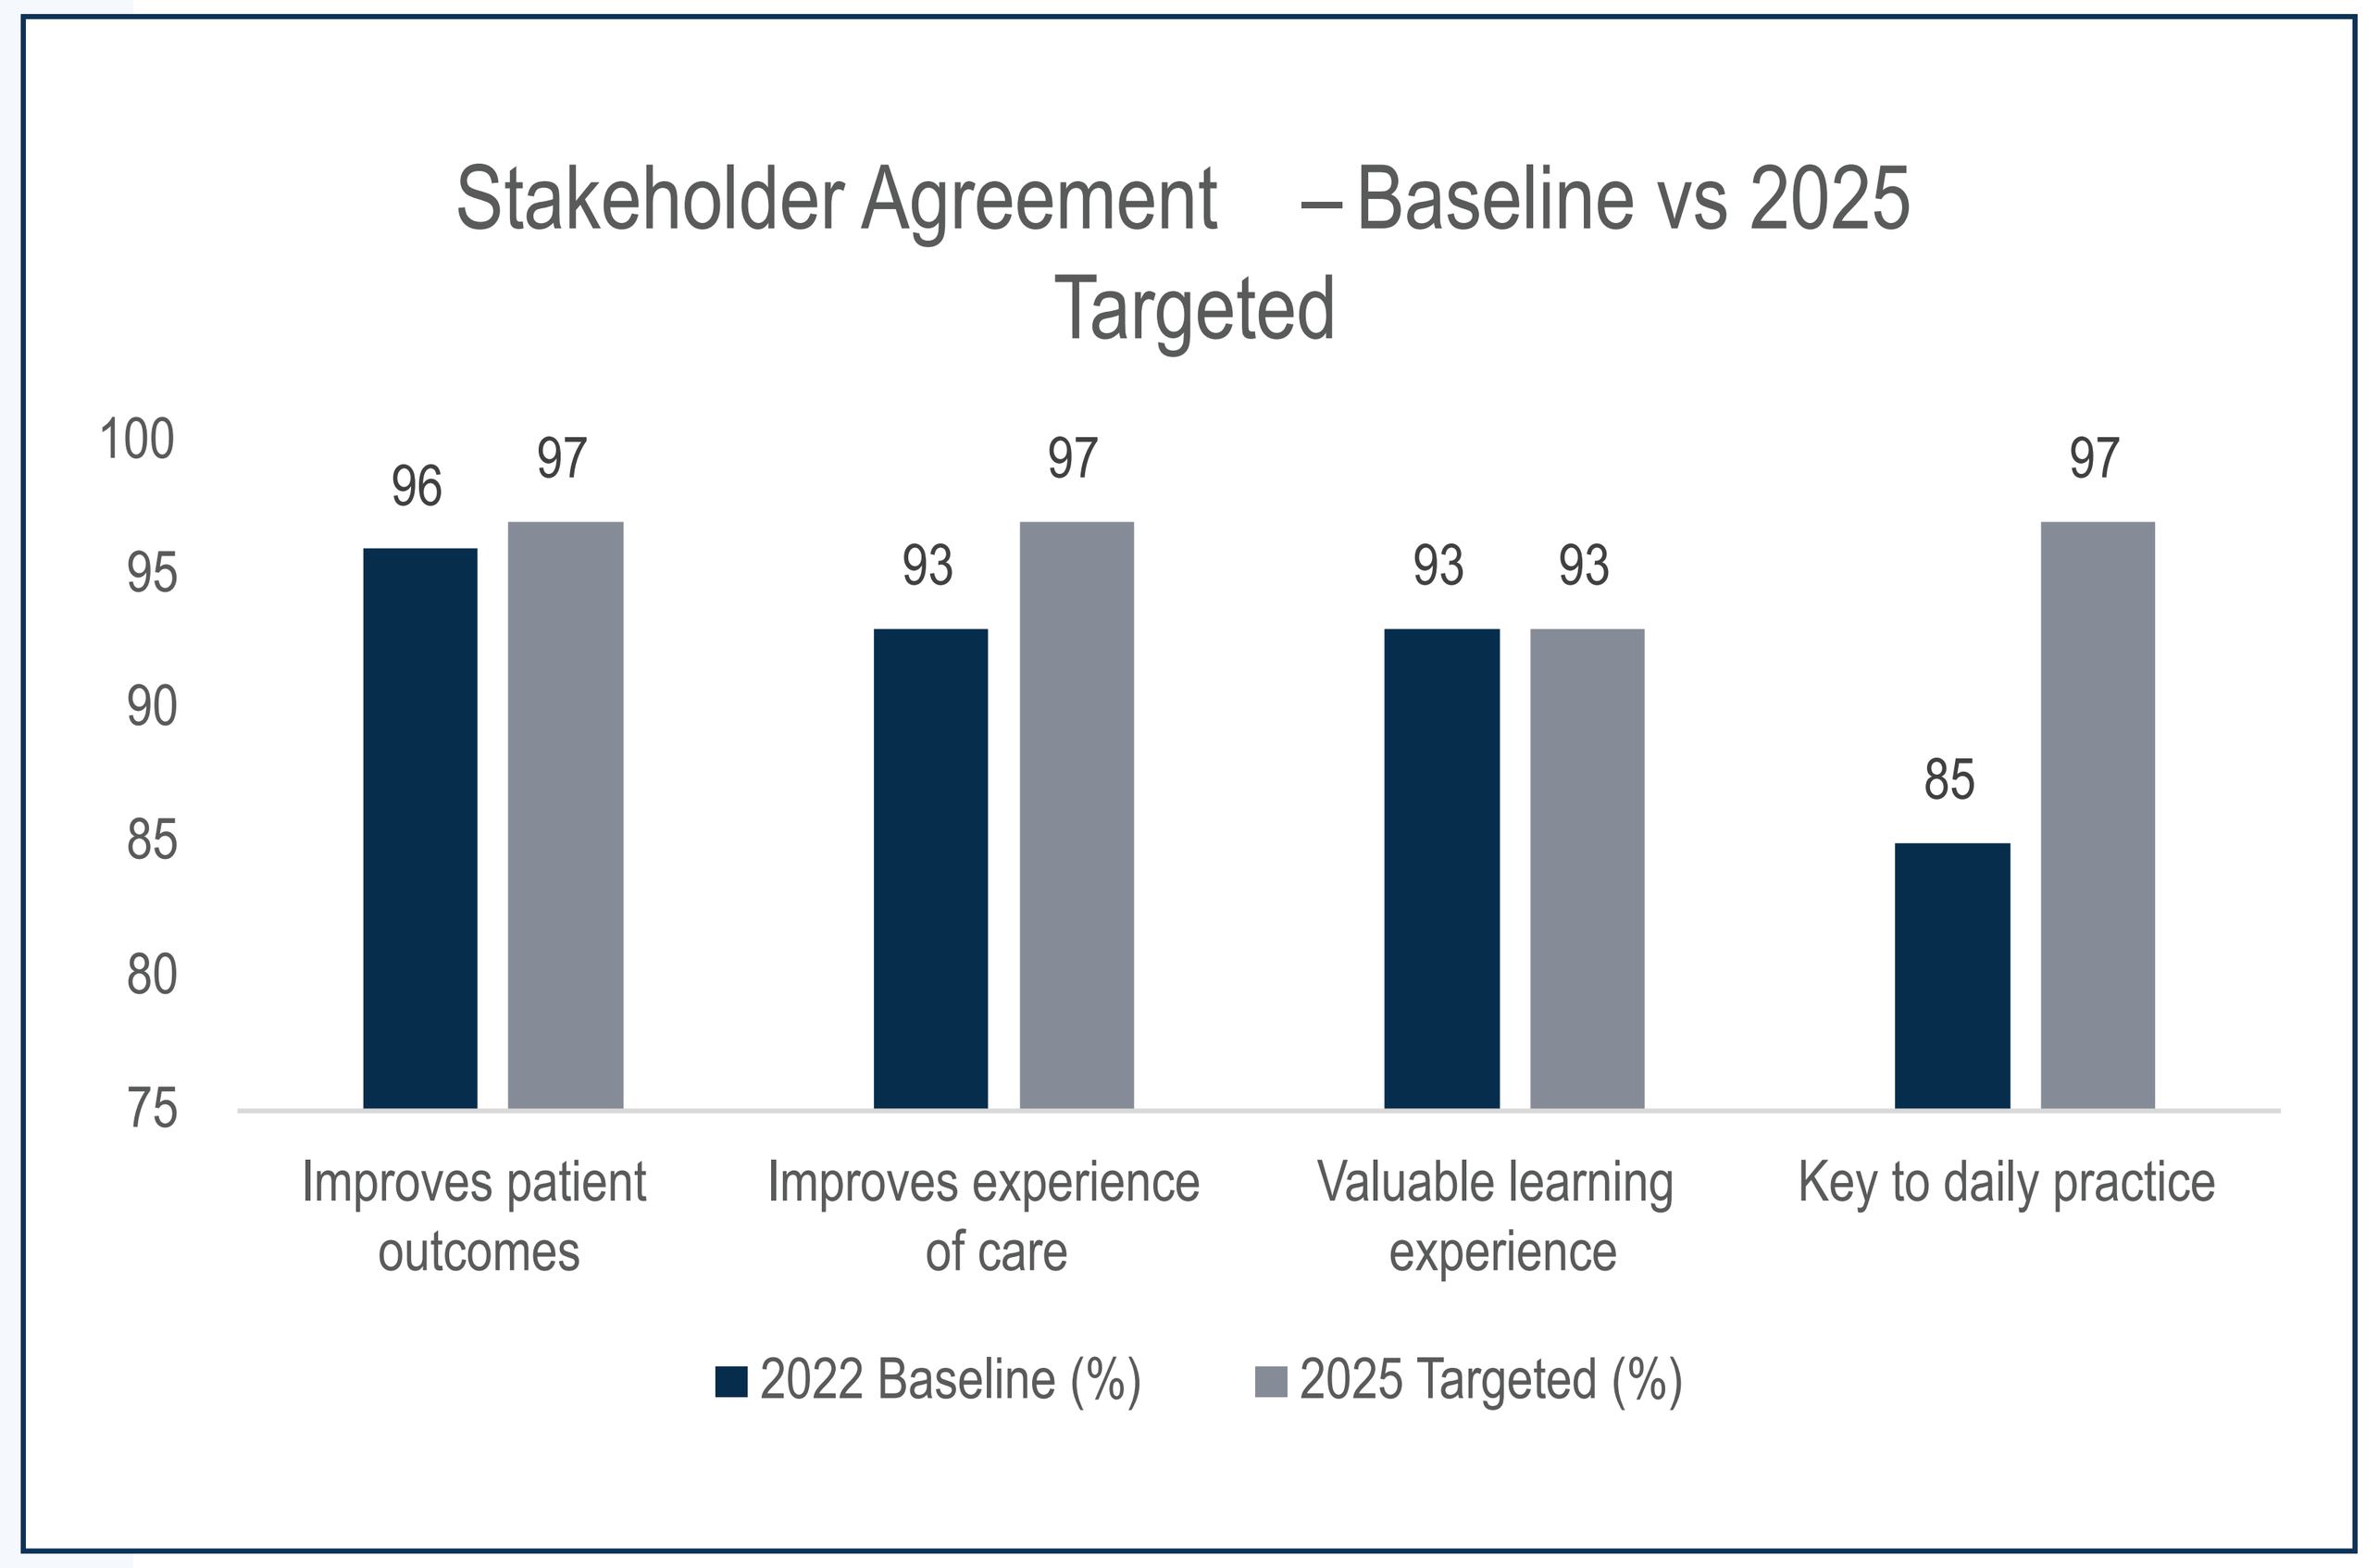

Stakeholder Agreement – Baseline vs 2025 Targeted
100
95
90
85
80
75
96
97
93
97
93
93
85
97
Improves patient outcomes
Improves experience of care
Valuable learning experience
Key to daily practice
2022 Baseline (%)
2025 Targeted (%)

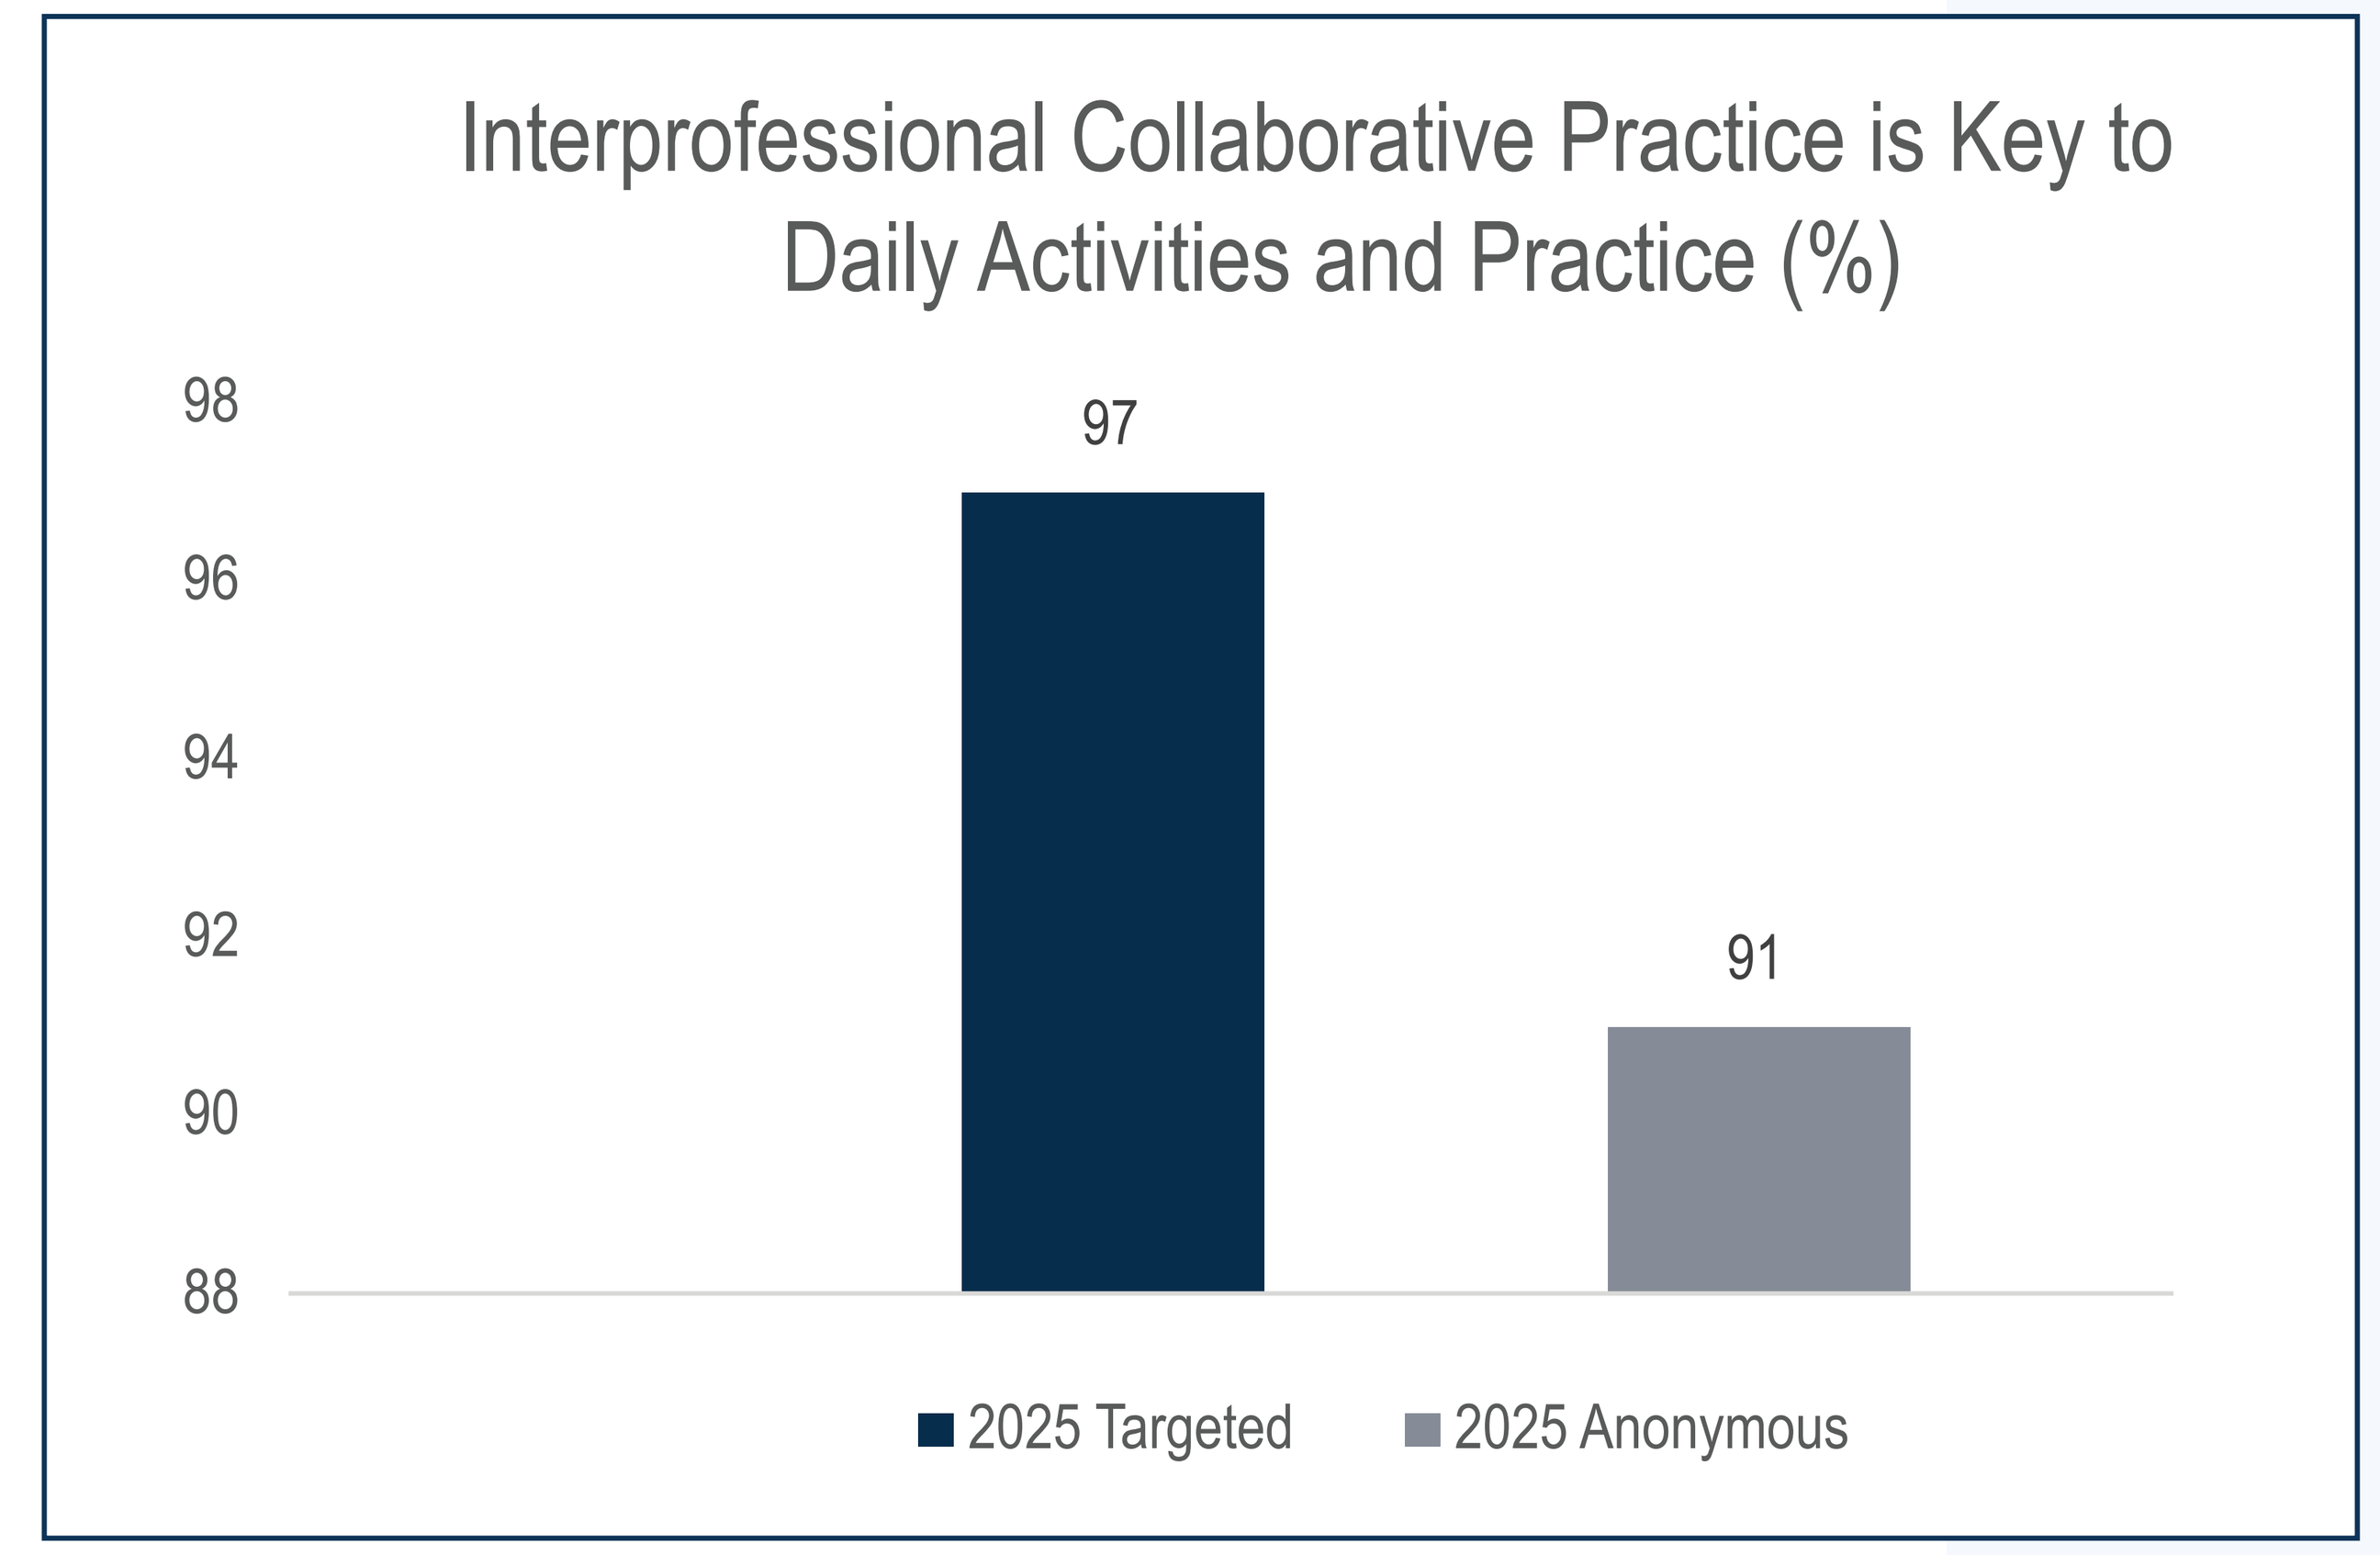

Interprofessional Collaborative Practice is Key to Daily Activities and Practice (%)
98
96
94
92
90
88
97
91
2025 Targeted
2025 Anonymous

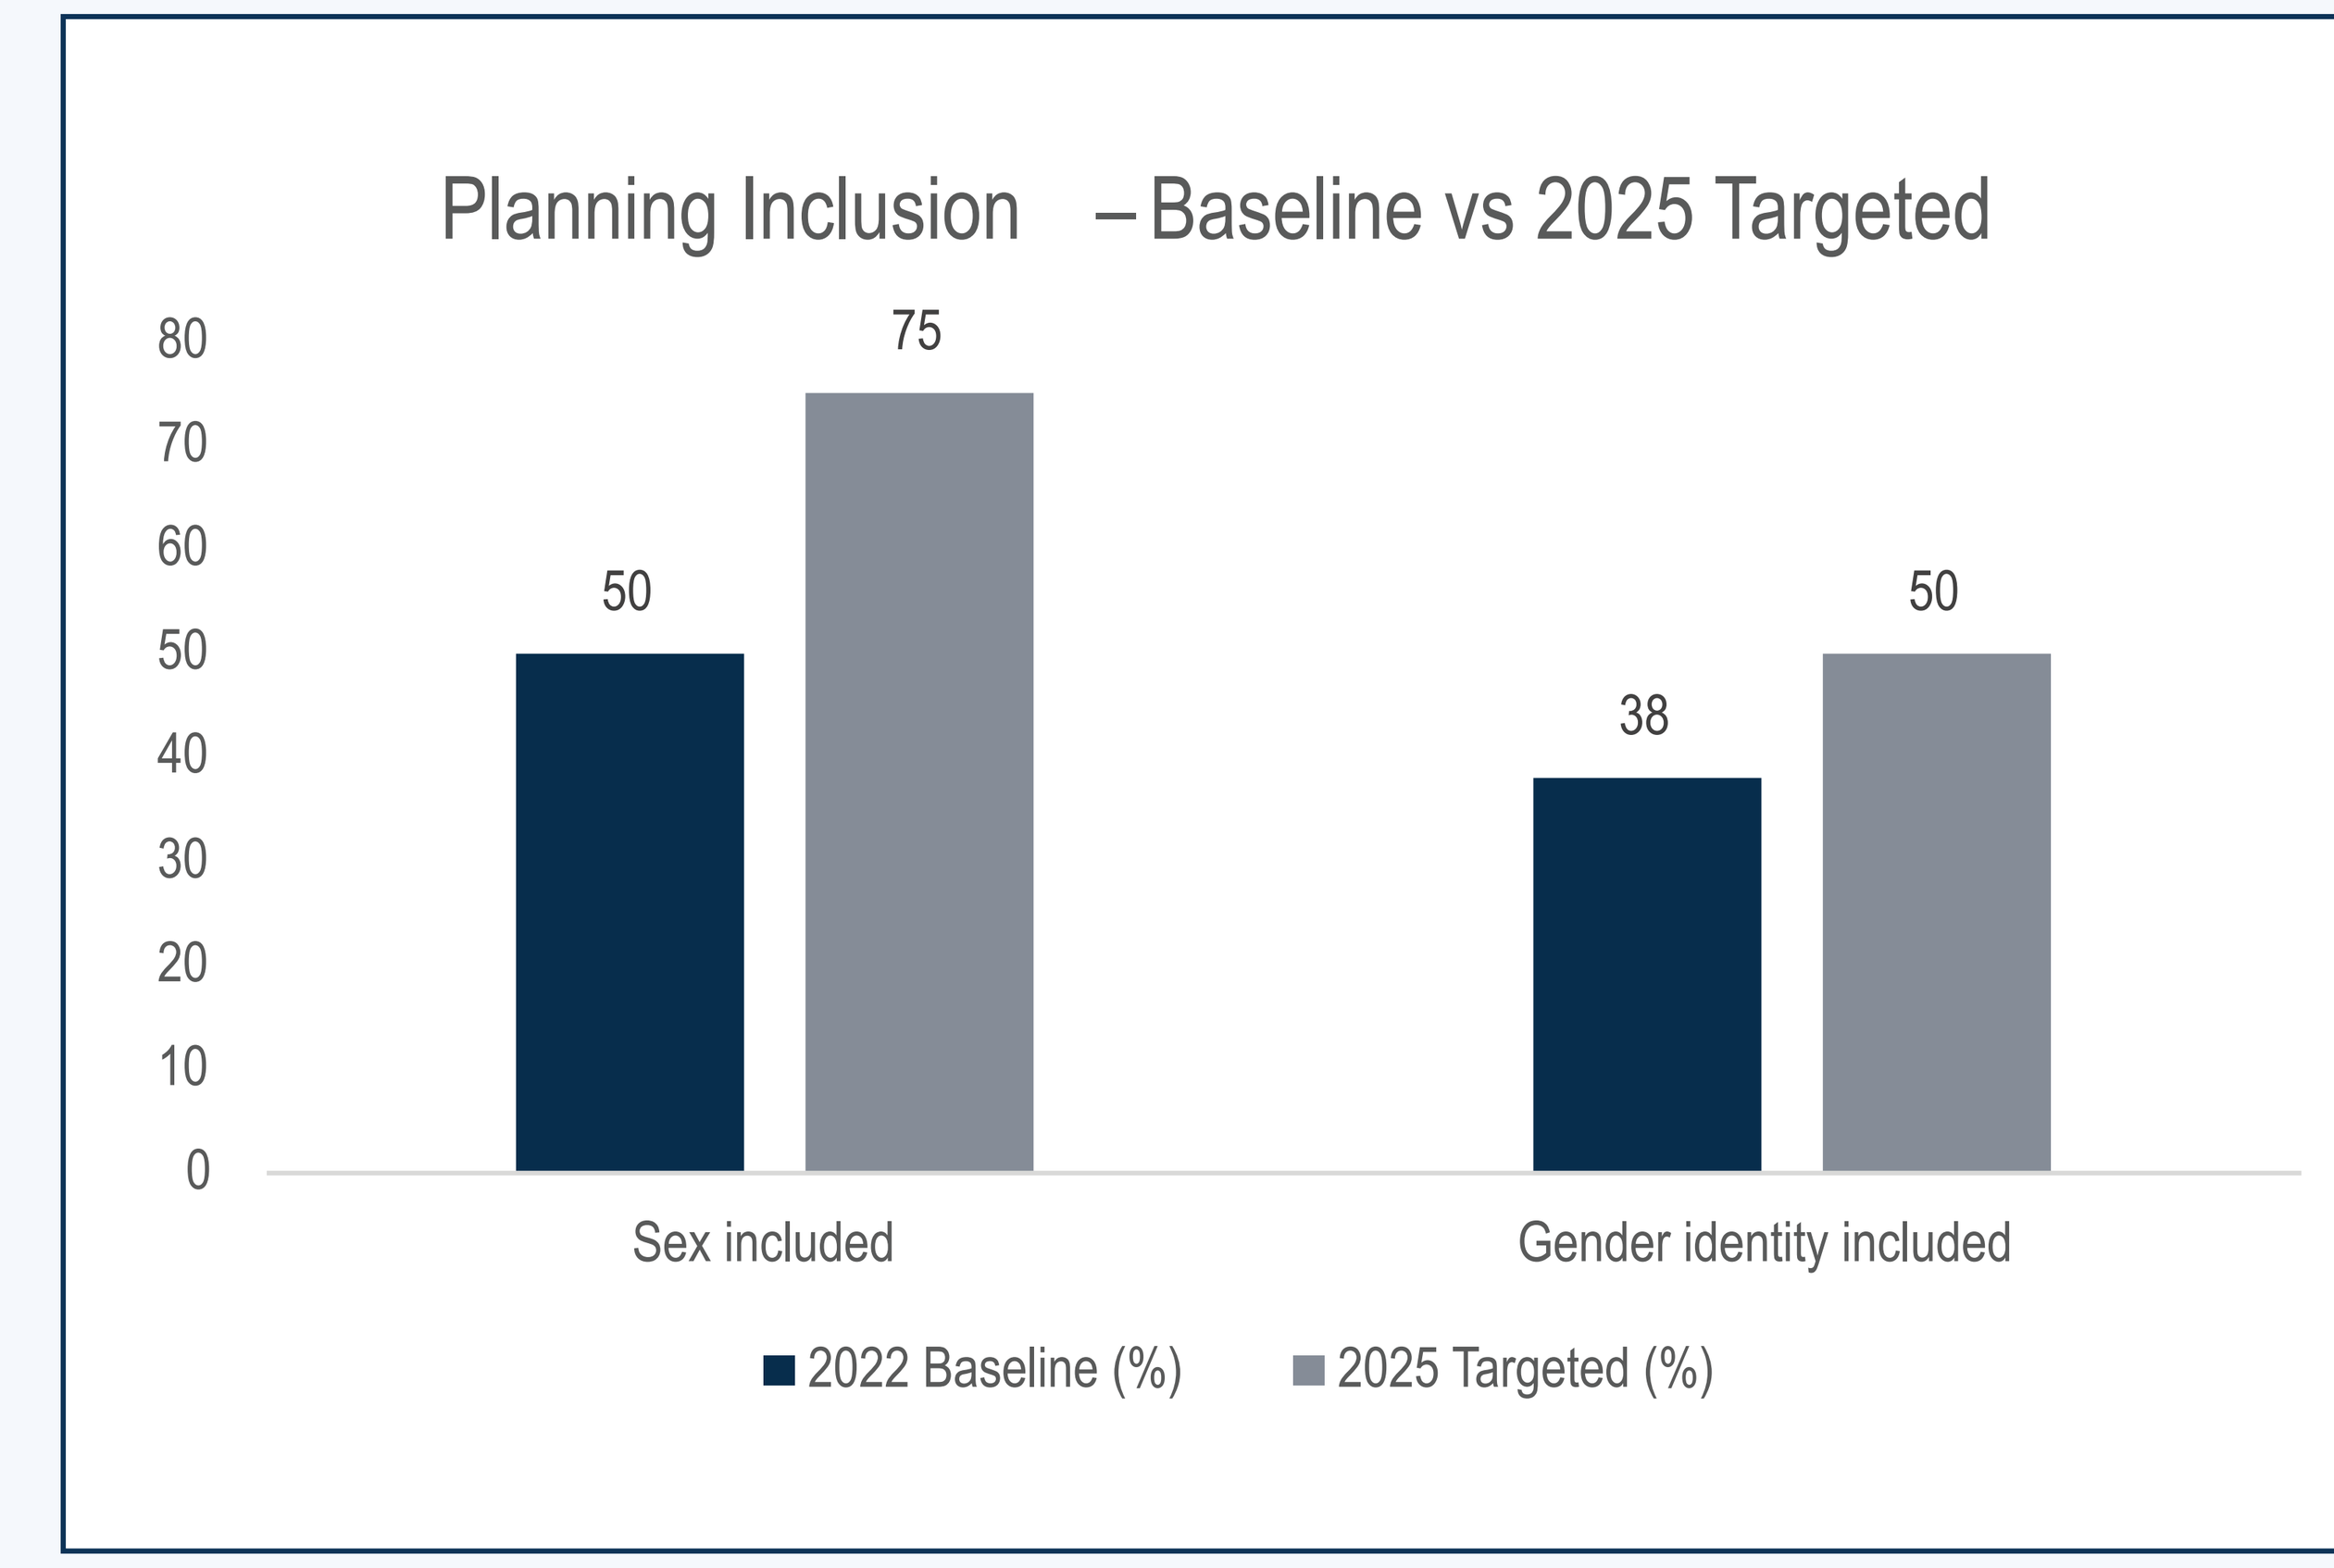

Planning Inclusion – Baseline vs 2025 Targeted
80
70
60
50
40
30
20
10
0
50
75
38
50
Sex included
Gender identity included
2022 Baseline (%)
2025 Targeted (%)

RESULTS

- **Valuing of interprofessional collaborative practice remained high between baseline and follow-up cohorts.**
 Agreement that interprofessional collaboration improves patient outcomes increased slightly from **96% at baseline to 97% in 2025**, while agreement that IPCP improves the experience of providing care increased from **93% to 97%**.

- **Interprofessional collaboration became more integrated into daily professional practice.**
 Agreement that IPCP is integral to daily professional activities increased from **85% at baseline to 97%** in the follow-up cohort.

- **Consideration of age, biological sex, and gender identity evolved within educational planning.**
 Among planning stakeholders, directional increases were observed in consideration of biological sex and gender identity during educational planning processes. Consideration of age demonstrated variability between cohorts. Interpretation is limited by smaller planning sample sizes.

- **Interprofessional concepts extended beyond formal planning stakeholders into broader professional environments.**
 Responses from the anonymous survey cohort demonstrated similarly high valuing of interprofessional collaboration, suggesting that IPCE concepts extend beyond formal continuing education leadership roles into broader professional interactions.

DISCUSSION

Findings demonstrate s**ustained high valuing of interprofessional collaborative practice** alongside **increased recognition of its relevance to daily professional activities**.

Results suggest movement beyond **reaction-level educational outcomes** toward **integration of interprofessional collaboration within organizational CPD culture**.

Evidence of similar perceptions among anonymously surveyed professionals indicates potential **diffusion of interprofessional concepts beyond structured planning environments**.

CONCLUSIONS

Longitudinal assessment demonstrates sustained normalization of interprofessional collaborative practice within an accredited CPD system, with increasing integration into professional identity and educational planning practices.

These findings support the role of **ongoing IPCE implementation and longitudinal evaluation** in advancing **organizational culture change and continuous improvement** aligned with Joint Accreditation expectations.

FUTURE DIRECTIONS

- **Continued longitudinal monitoring** of stakeholder perceptions
- **Expanded faculty development** supporting equity-informed planning
- **Evaluation of behavioral and practice-level outcomes** associated with IPCE participation

ACKNOWLEDGMENTS

University of Kansas Medical Center

No commercial funding was received for this project.

All authors report no relevant financial

KU MEDICAL CENTER
The University of Kansas

KELLY SABOL*
MOLLY SMITH, MS**
KIM TEMPLETON, MD, FAAOS, FAOA, FAMWA***
TERI KENNEDY, PHD, MSW, ACSW, FGSA, FNAP****

*KUMC Area Health Education Center; **KUMC Office of Continuing Education and Professional Development; ***University of Kansas School of Medicine; ****University of Kansas School of Nursing; The authors and contributors report no relationships of a financial, professional or personal nature.

UNIVERSITY OF KANSAS MEDICAL CENTER
WWW.KUMC.EDU